The impact of unmodelled interoccasion variability in bioavailability and absorption on parameter estimates in population pharmacokinetic analysis

S. Koehne-Voss (1), A. Gautier (1), G. Graham (1)

(1) Novartis Pharma AG Basel

Background and Objectives

Background: Karlsson and Sheiner [1] showed that ignoring interoccasion variability (IOV) in pharmacokinetic data can lead to biased population pharmacokinetic parameter estimates. They proposed a model that incorporates IOV. Karlsson and Sheiner only considered models with intravenous administration in their simulations, so did not study the effect of unmodelled IOV in bioavailability and absorption parameters.

Objectives: We study the effect of interoccasion variability in bioavailability and absorption that is present in the data but not accounted for in the population pharmacokinetic analysis model on parameter estimates.

Methods

Simulations were performed. Data were generated from a one-compartment model with first-order absorption. The model included independent (diagonal) random effects for absorption (ka), apparent clearance and volume (CL/F, V/F), and bioavailability (F). Parameter values were V/F = 1, CL/F = 0.1, ka = 0.2, F = 1 (F fixed for estimation). Between subject standard deviations were 0.3, i.e. $\omega_{V/F} = \omega_{CL/F} = \omega_{ka} = \omega_F = 0.3$. Residual error (exponential error model) was $\omega_{RES} = 0.3$. IOV between two occasions was present either in relative bioavailability (F) or absorption rate (ka).

Two study designs were considered. In the first design single dose data were generated for occasion 1 and steady state data for occasion 2 (SD/MD design). In the second design steady state data were simulated on both occasions (MD/MD design). Dense (20 subjects with 12 samples each) and sparse (60 subjects with 4 samples each) sampling strategies were considered. In total 8 different scenarios were simulated by combining 2 sources of IOV (ka or F), 2 designs (SD/MD, MD/MD), and 2 sampling schemes (rich, sparse).

In each scenario, simulations were performed with either small ($\omega_{IOV} = 0.15$), medium ($\omega_{IOV} = 0.30$), or large IOV ($\omega_{IOV} = 0.50$). N = 250 studies were simulated for each scenario and value of IOV.

Simulated datasets were analyzed with NONMEM V7.3 using the FOCEI method and an analysis model that matched the data generating model or an analysis model that matched the data generating model but with interoccasion variability in ka or F not accounted for.

Results

- IOV in ka

Figure 1 shows the bias in the parameter estimates when IOV in ka is accounted for (left panel) or not accounted for in the analysis model (right panel). A scenario with a single dose administration at the first occasion and steady state data at the second occasion (SD/MD design) and sparse sampling is shown.

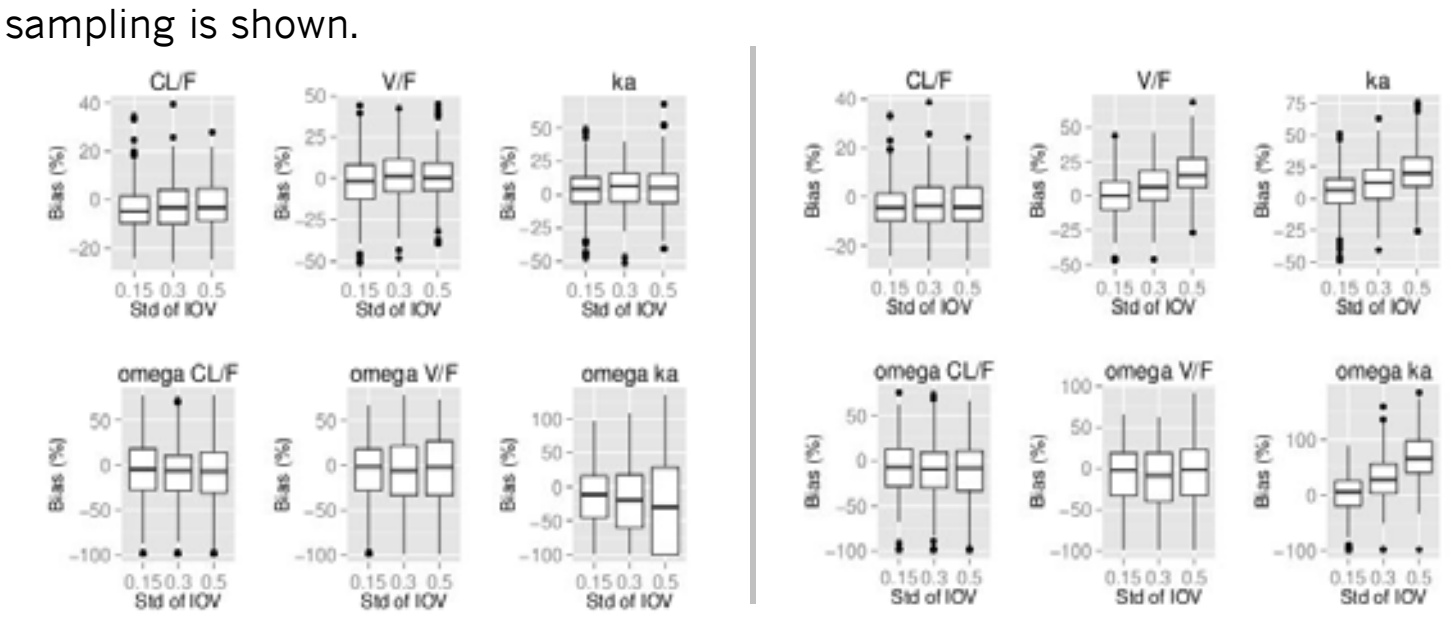
CL/F
V/F
ka
omega CL/F
omega V/F
omega ka
Bias (%)
Std of IOV

Figure 1: Bias in parameter estimates when IOV in ka is accounted for (left panel) or not accounted for in analysis model (right panel); SD/MD design, sparse sampling

Figure 1 shows that when IOV in ka is not accounted for in the population PK model there is positive bias in the estimates of V/F, ka, and ω_{ka}. The bias increases with the magnitude of the IOV and is not seen when IOV is incorporated into the model. While the median bias for V/F and ka is <25% even with large values of IOV ($\omega_{IOV} = 0.50$), the median bias ω_{ka} is up to ca. 70% with large IOV.

In terms of bias estimates of CL/F seem to be less affected by unmodeled IOV in ka. Similar results were seen with rich sampling and MD/MD designs.

- IOV in F

Figure 2 shows the bias in the parameters estimates when IOV in F is accounted for (left panel) or not accounted for in the analysis model (right panel). A scenario with a single dose administration at the first occasion and steady state data at the second occasion (SD/MD design) and sparse sampling is shown.

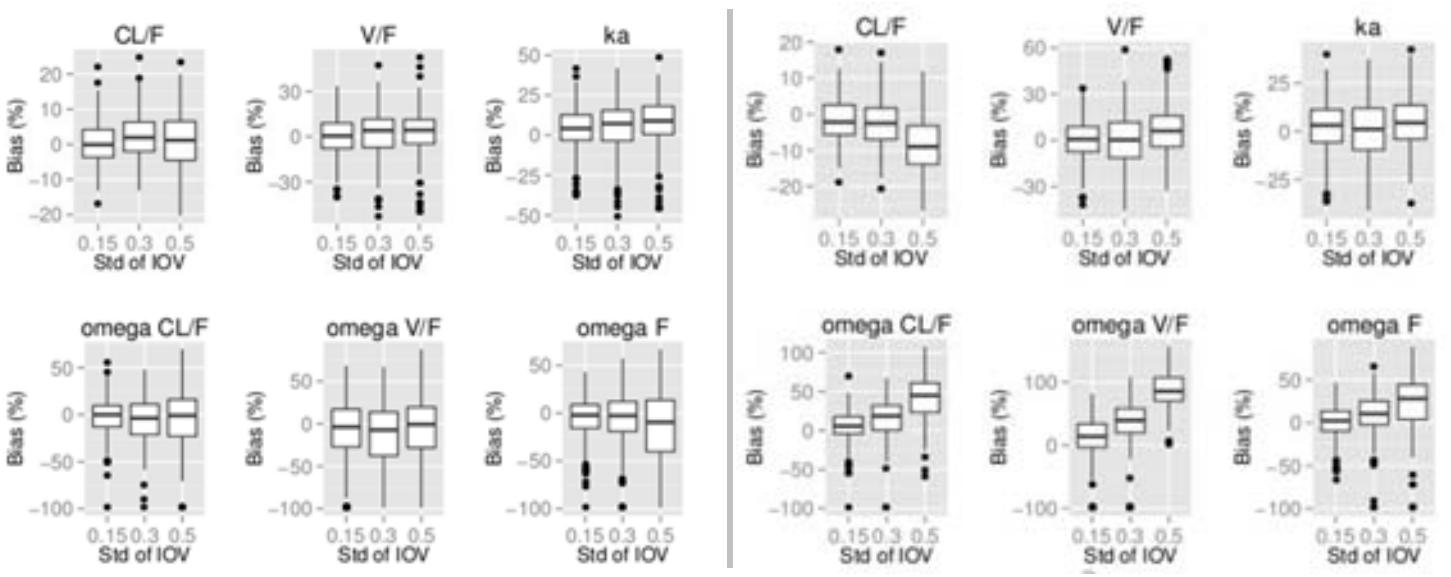
CL/F
V/F
ka
omega CL/F
omega V/F
omega F
Bias (%)
Std of IOV

Figure 2: Bias in parameter estimates when IOV in F is accounted for (left panel) or not accounted for in analysis model (right panel); SD/MD design, sparse sampling

Figure 2 shows that when IOV in F is not accounted for in the SD/MD design there is a clear trend to overestimate $\omega_{CL/F}$, $\omega_{V/F}$, and ω_F. Bias can be subtantial. E.g. for $\omega_{V/F}$ median bias is ca. 90% with large IOV.

In terms of bias estimates of fixed effect parameters (CL/F, V/F, ka) seem to be less affected by unmodeled IOV in F and show only small bias. Results with rich sampling are similar.

Figure 3 shows that the bias in $\omega_{V/F}$ and $\omega_{CL/F}$ which is seen in the SD/MD design, is not observed in the MD/MD design while the strong bias for ω_F is maintained.

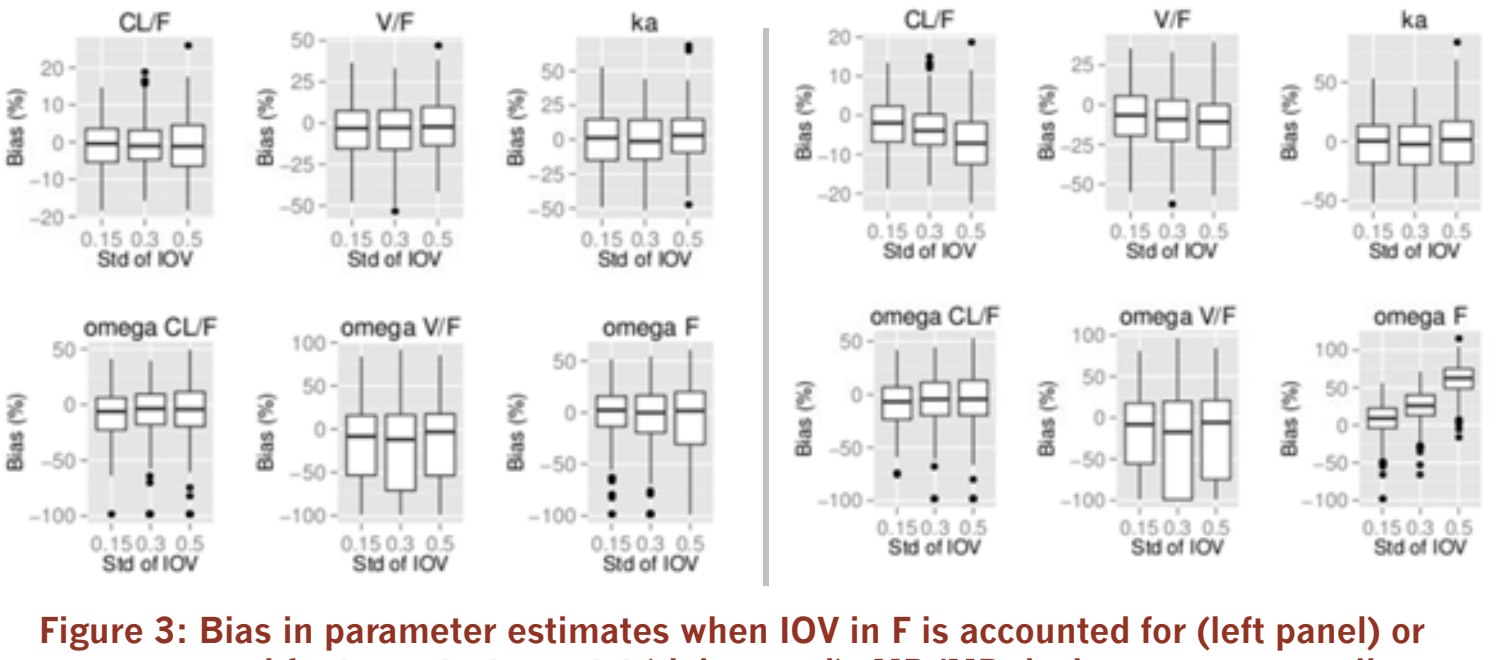
CL/F
V/F
ka
omega CL/F
omega V/F
omega F
Bias (%)
Std of IOV

Figure 3: Bias in parameter estimates when IOV in F is accounted for (left panel) or not accounted for in analysis model (right panel); MD/MD design, sparse sampling

For both ka and F, estimates of residual error were inflated when IOV is not incorporated into the model.

Conclusions

When sampling pharmacokinetic data on several occasions interoccasion variability in absorption and bioavailability should be included in the model to avoid potential bias in population pharmacokinetic parameter estimates. In this simulation example bias was more pronounced in random effect parameters compared to fixed effect parameters.

References

1. Karlsson MO, Sheiner LB. The importance of modeling interoccasion variability in population pharmacokinetic analyses. J Pharmacokinet Biopharm (1993) 21(6):735-50

PAGE meeting – June 2015

NOVARTIS PHARMACEUTICALS